Preliminary study of efficacy and duration of an mRNA based GLP-1R agonist in diabetic monkeys

Fengwei Xu, KangZeng Wu, Lei Zhang, Yongchao Dai, Yuhong Xu

Hangzhou HighField Biopharmaceuticals. www.highfieldbio.com

BACKGROUND

HFG1 is an mRNA based GLP-1R agonist encoding a fusion protein of a GLP-1 analogue and a pH sensitive binder of human FcRn. The mRNA is encapsulated inside lipid nano-structures (LNPs) designed for subcutaneous gene delivery and local transfection, and the proteins expressed would be secreted and distributed into systemic circulation. In order to characterize the HFG1 activities in vivo, five monkeys with naturally occurred diabetes (FPG≥9.69 mg/dl, HbA1c≥5.6%) were selected to receive one subcutaneous injection of LNPs of slightly variable compositions containing about 0.5-1.0mg mRNA. The monkeys were closely monitored for 60 days and no behavior changes were observed except that they all had greatly reduced food uptake on the first few days after injection. By D56 they lost an average of 0.22kg each. Blood samples were collected at various time points and assayed in a GLP-1R/CRE Luciferase Reporter Cell system for GLP-1 equivalent activities. Figure 1 plotted the individual measurements (solid dots) and average value changes (solid line). The HbA1c% levels were assayed by Suzhou Xihua Scientific Co. and shown as open squares. Even though some time points are missing (TBD), the trend of HbA1c% reduction after a single HFG1 injection is significant.

METHODS

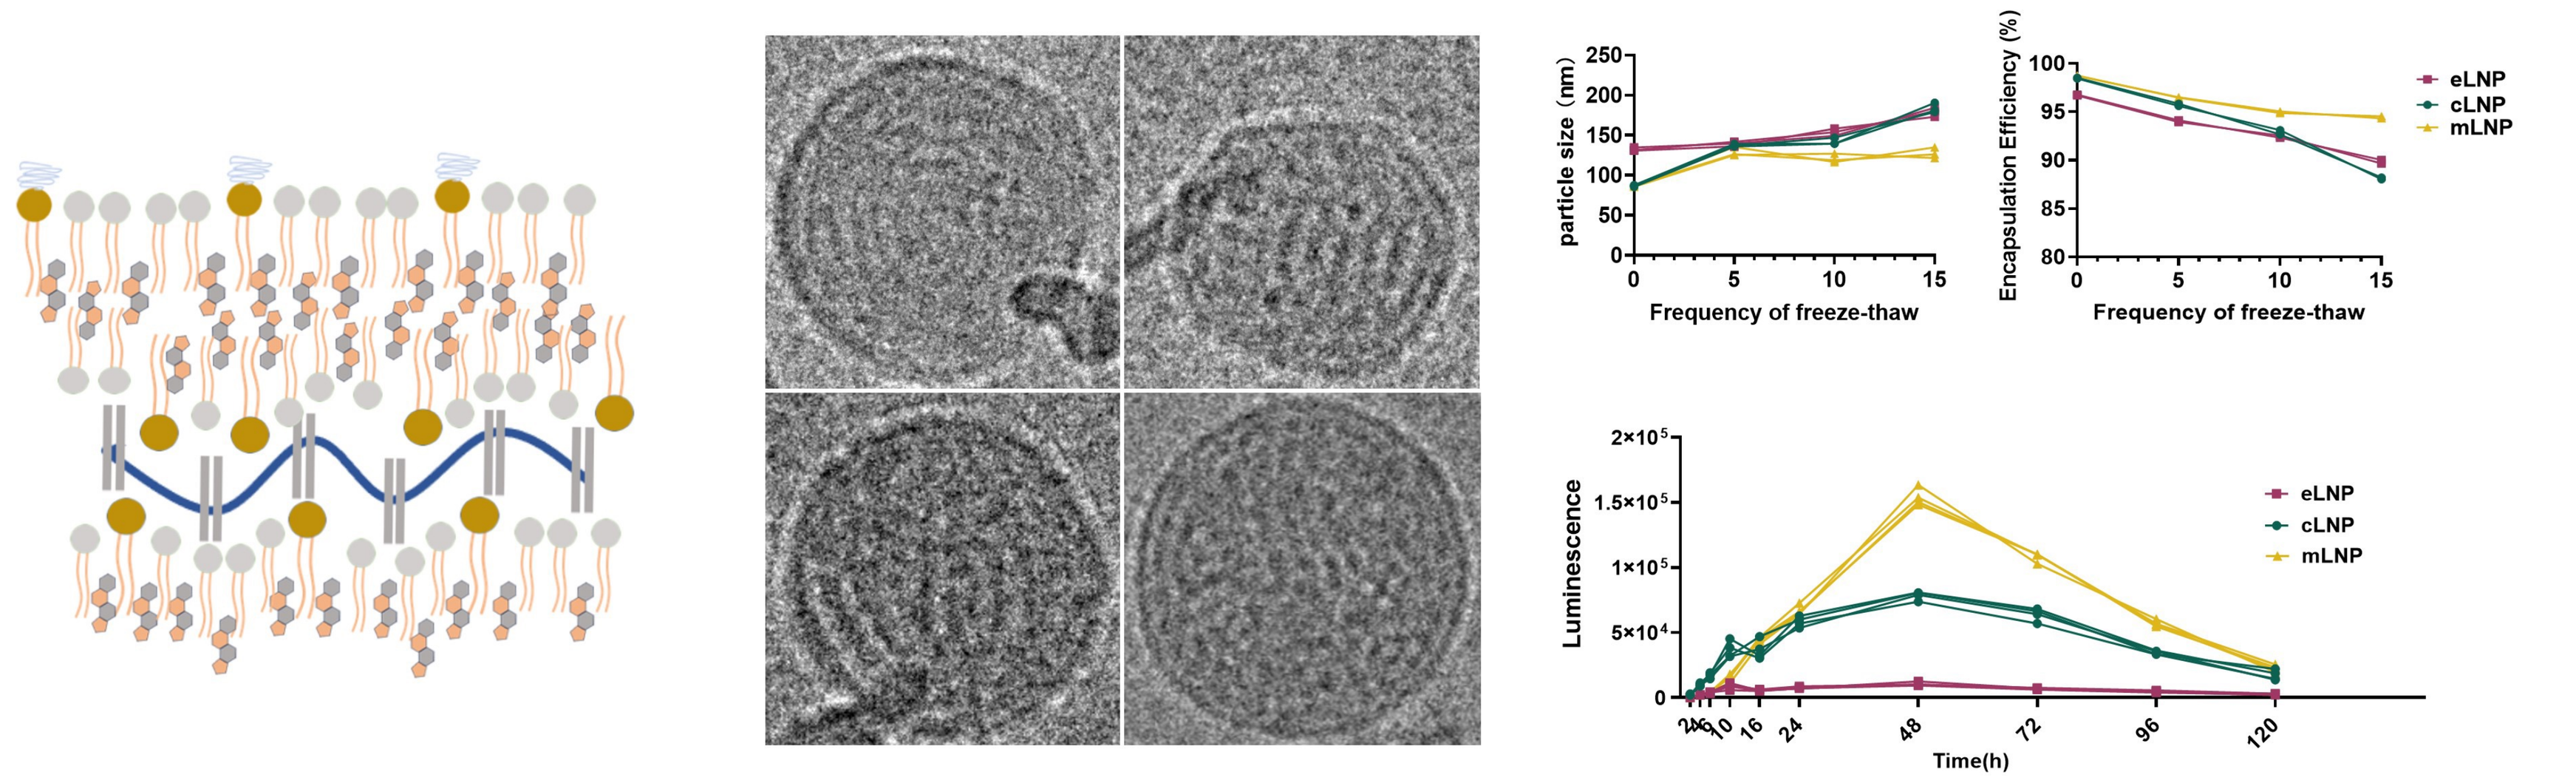
particle size (nm)
Frequency of freeze-thaw
Encapsulation Efficiency (%)
Frequency of freeze-thaw
eLNP
cLNP
mLNP
Luminescence
Time(h)

Figure 1. LNPs with membrane stabilized structures (mLNP) were developed. Unlike traditional LNPs for mRNA vaccination, mLNPs are more stable both under storage stress conditions and in physiological medium. The resulted transgene production is higher and longer lasting compared to other LNPs.

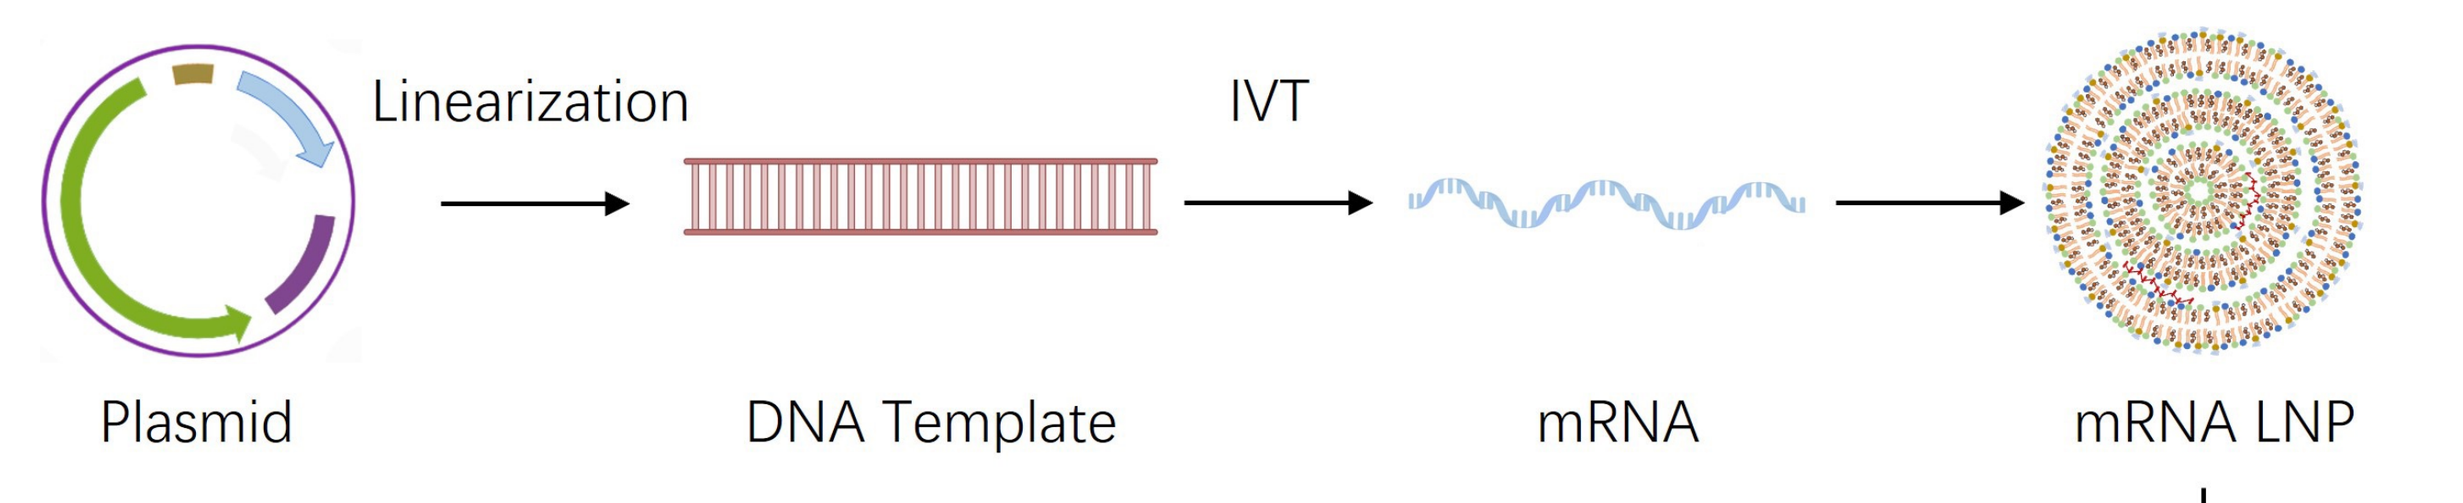
Linearization
IVT
Plasmid
DNA Template
mRNA
mRNA LNP

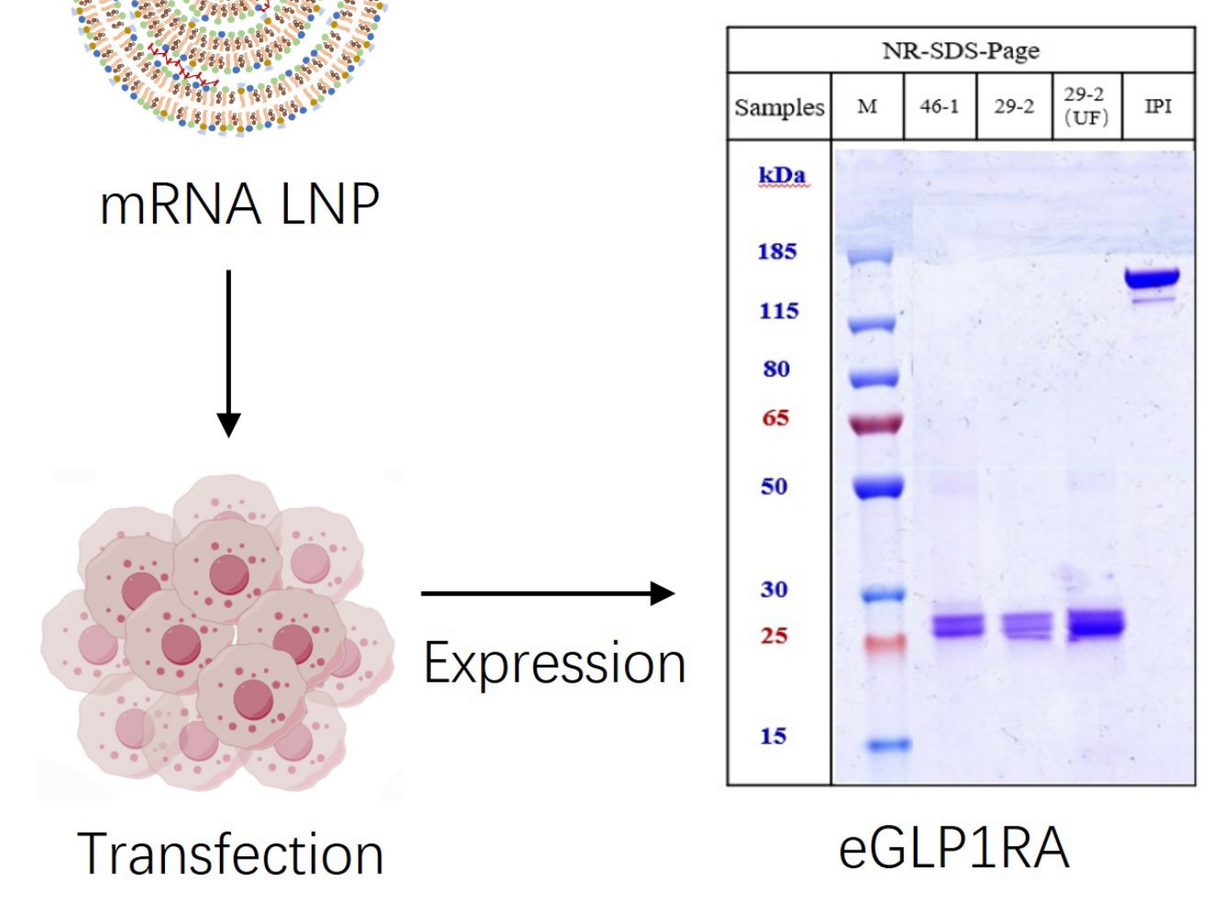
Transfection
Expression
NR-SDS-Page
Samples
M
46-1
29-2
29-2 (UF)
IPI
kDa
eGLP1RA

Figure 2. mRNA encoding the eGLP1RA sequence was made by in vitro transcription and packaged into a mLNP. These LNPs are to be tested in cell and animal models for their GLP1RA activities.

PRELIMINARY RESULTS

GLP1RA activities of the mRNA LNP transfection products

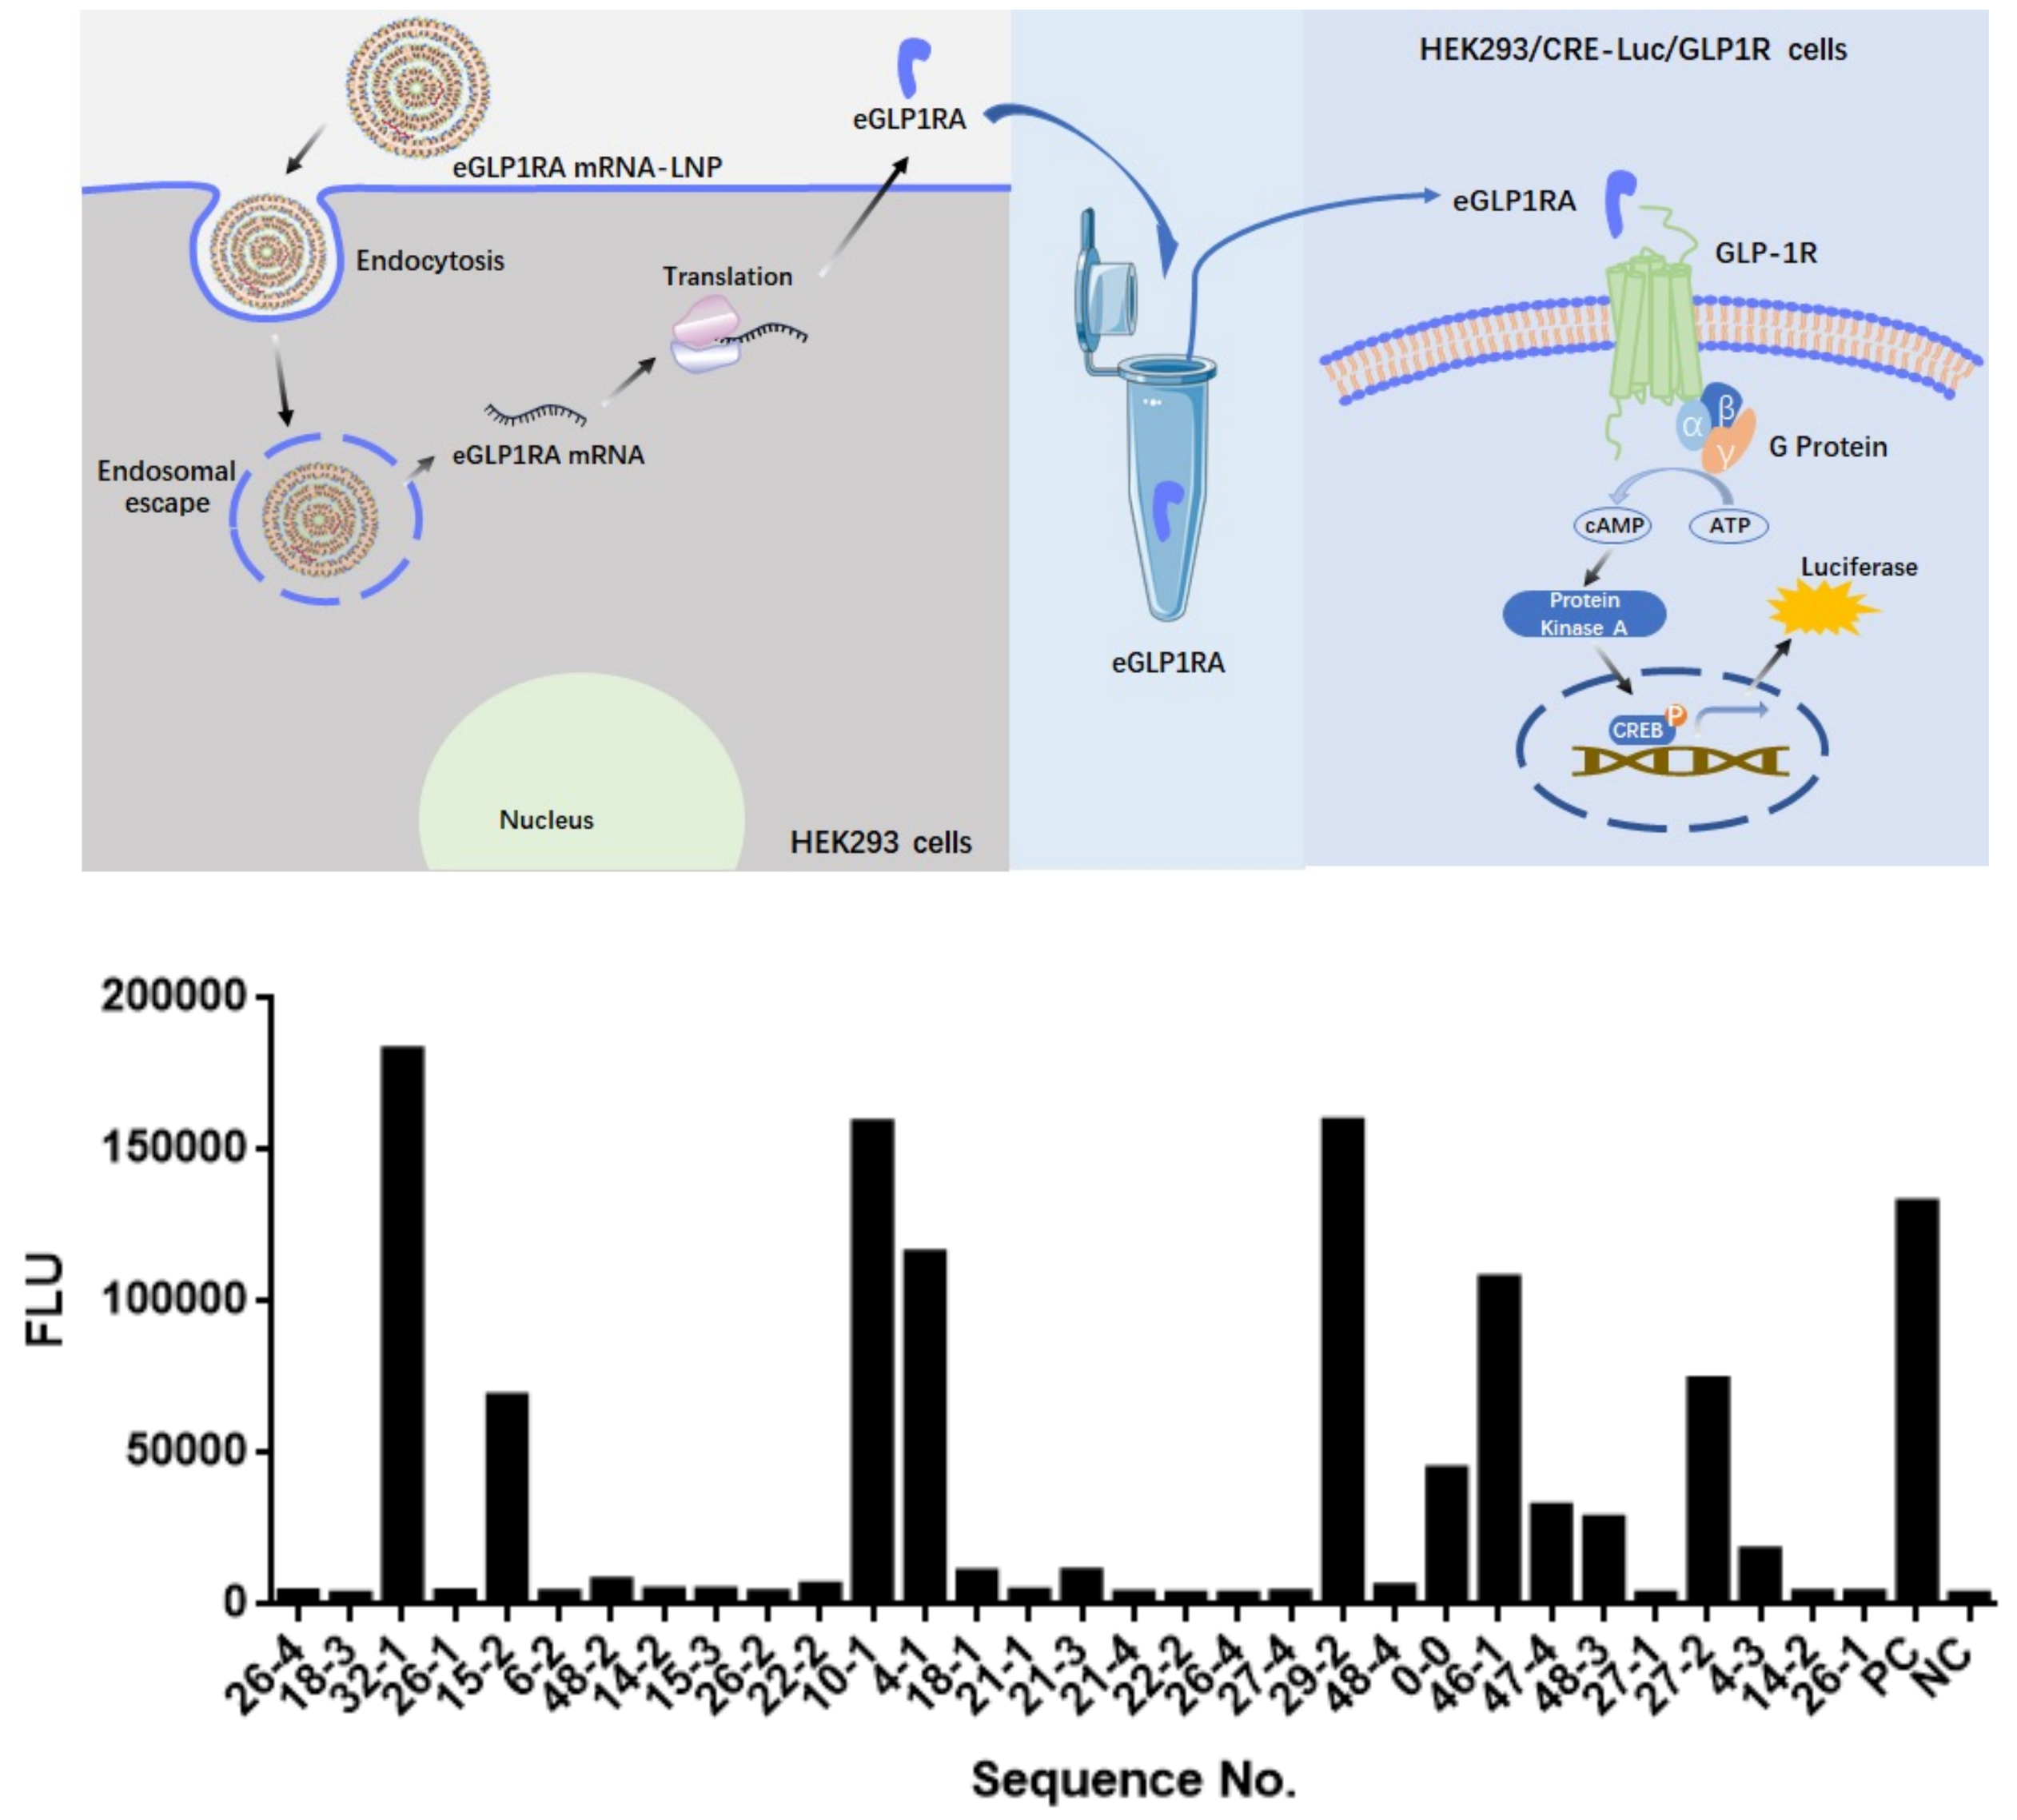
eGLP1RA mRNA-LNP
eGLP1RA
Endocytosis
Translation
Endosomal escape
eGLP1RA mRNA
Nucleus
HEK293 cells
HEK293/CRE-Luc/GLP1R cells
GLP-1R
G Protein
cAMP
ATP
Protein Kinase A
Luciferase
CREB
FLU
Sequence No.

Figure 3. The various mRNA sequences were used to transfect HEK293 cells using mLNPs. The resulted gene products were tested for GLP1RA activities using a HEK293/CRE-Luc/GLP1R assay. The sequences with the highest activities were selected for further development.

Kinetics of mLNPs and the gene products after s.c. injection

Figure 4. The mLNPs were administered by s.c. injection. There were two dose groups containing 3 monkeys each. G1-G3 received a 100ug mRNA dose (green dots and line) and G4-G6 received 300ug (blue dots and line). The plasma cationic lipid concentration (top figure), mRNA concentration (middle figure), and estimated eGLP1RA concentration (bottom figure) at various time points after injection were assayed and plotted. (Y axis marks are relative scale numbers that have not yet been validated.)

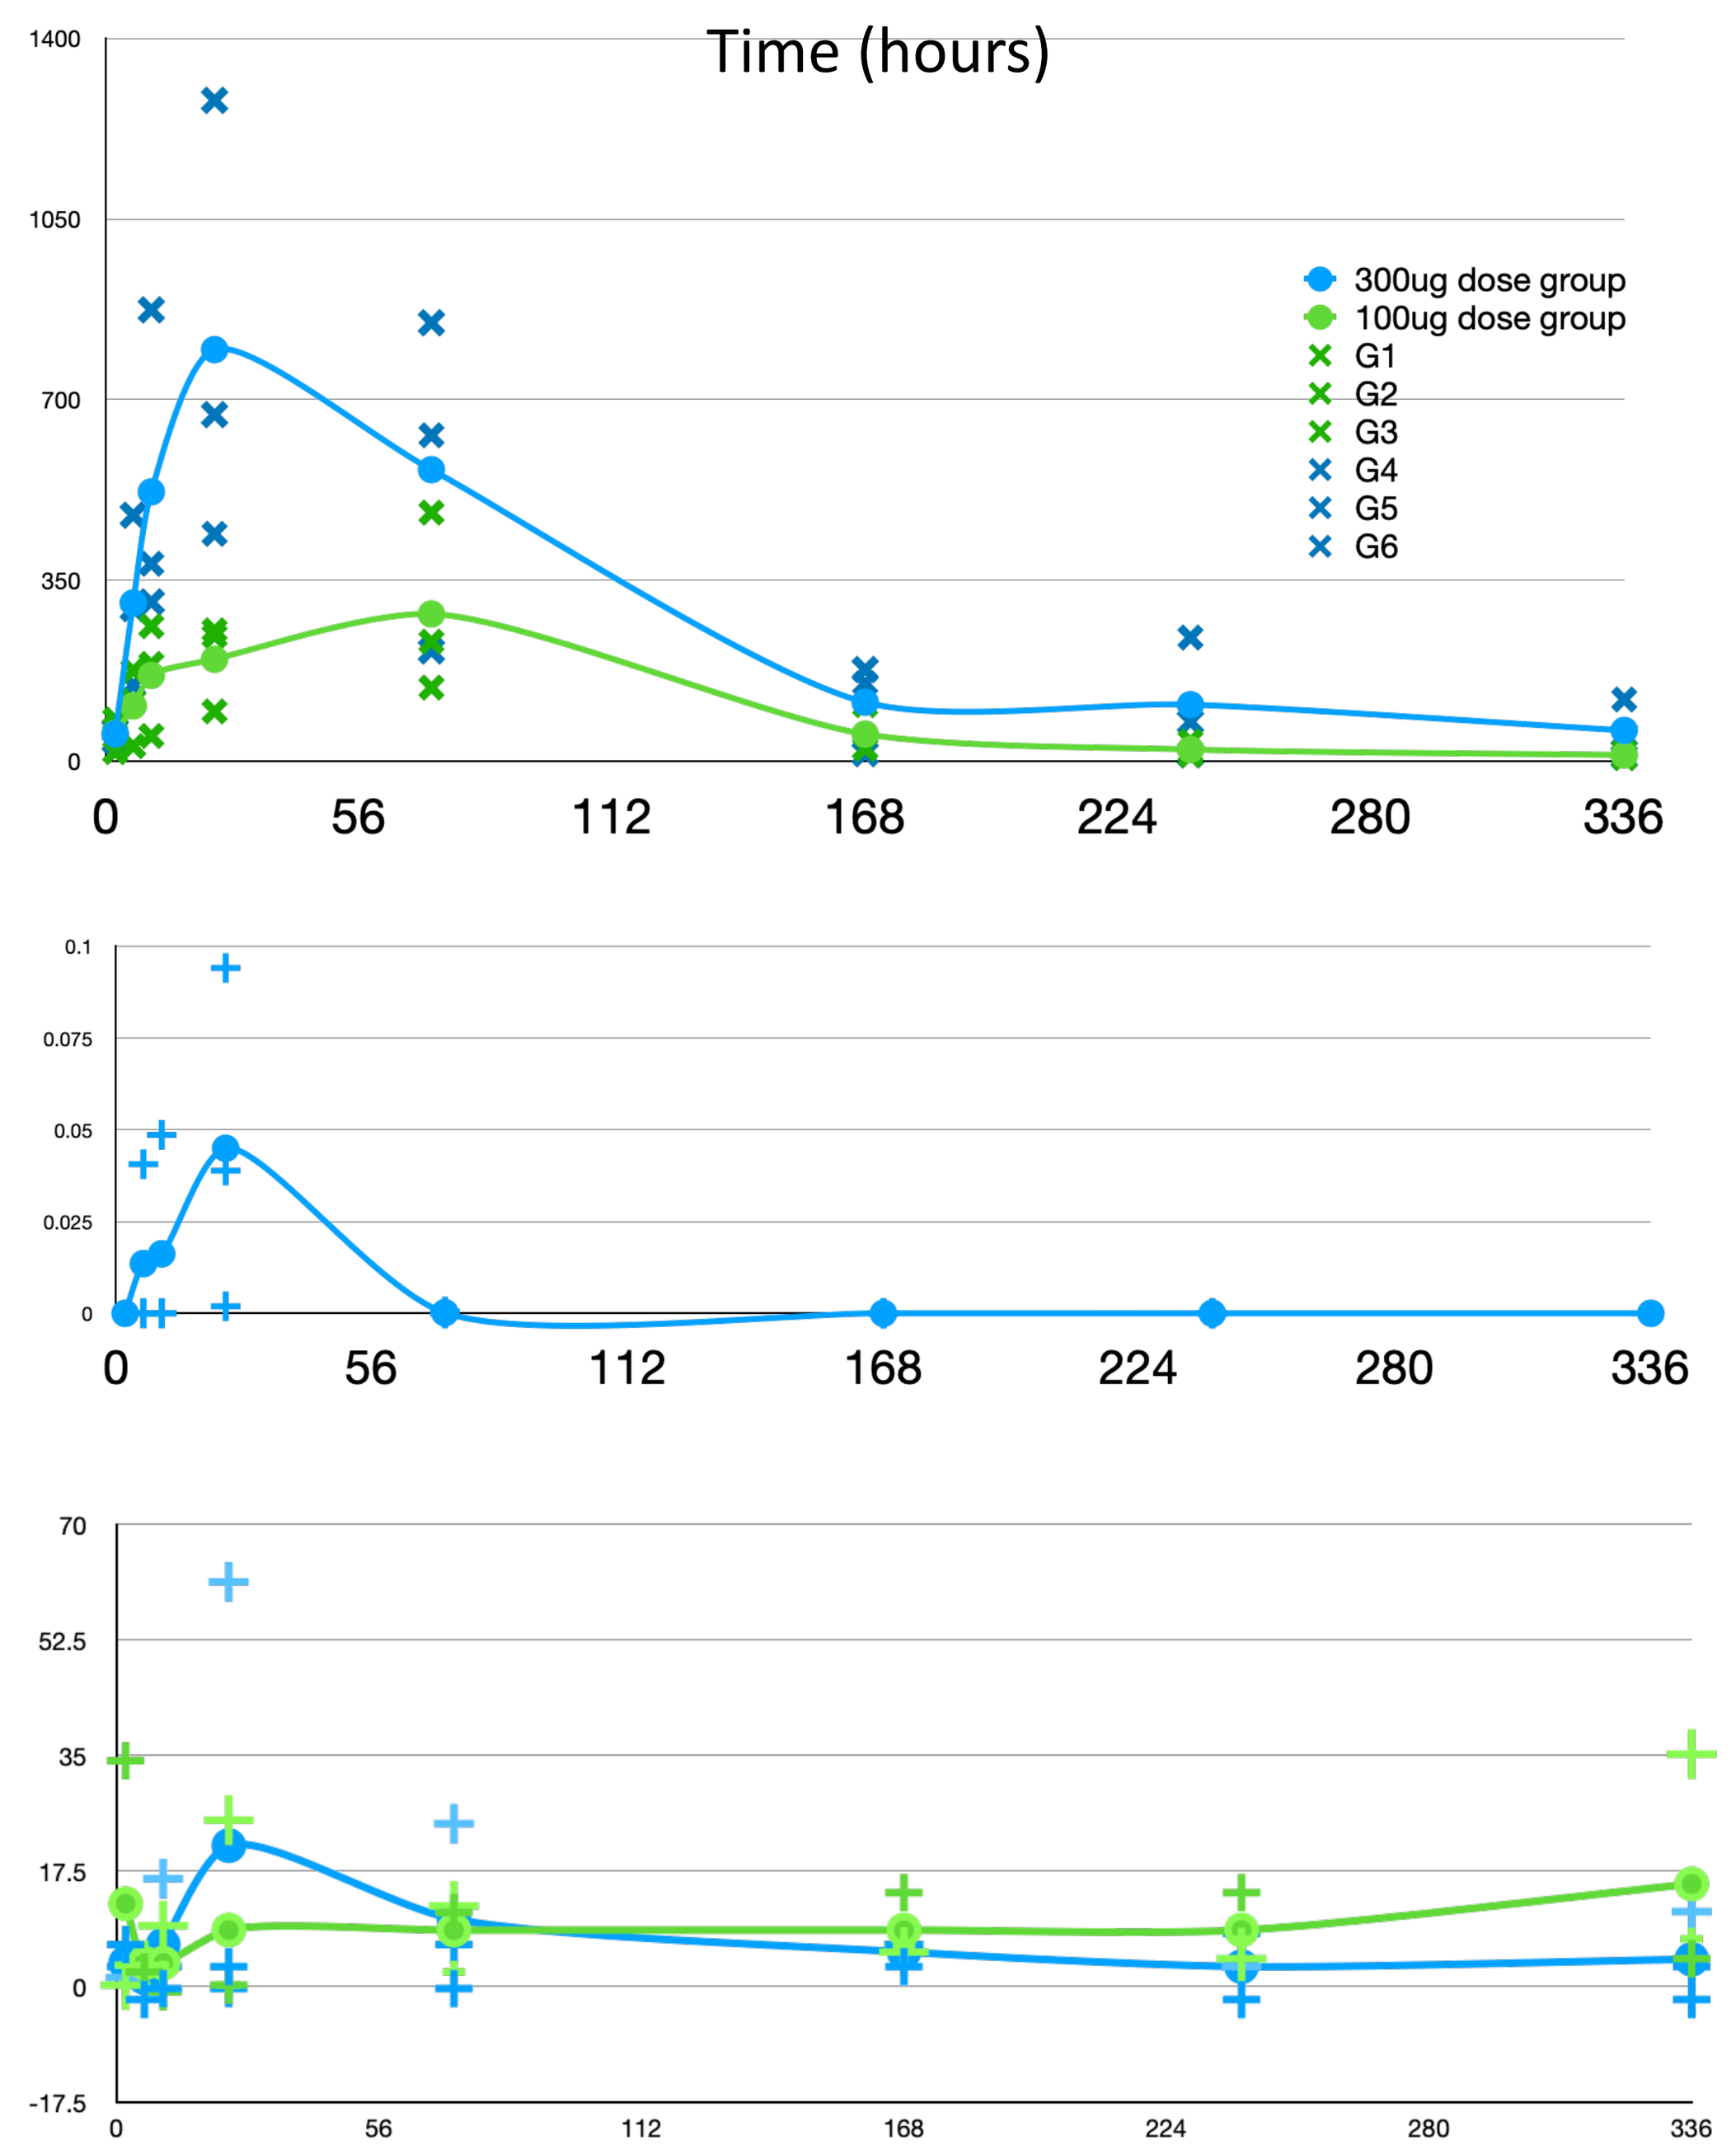
Time (hours)
300ug dose group
100ug dose group
G1
G2
G3
G4
G5
G6

Longer term PK/PD observations after a single HFG1 injection

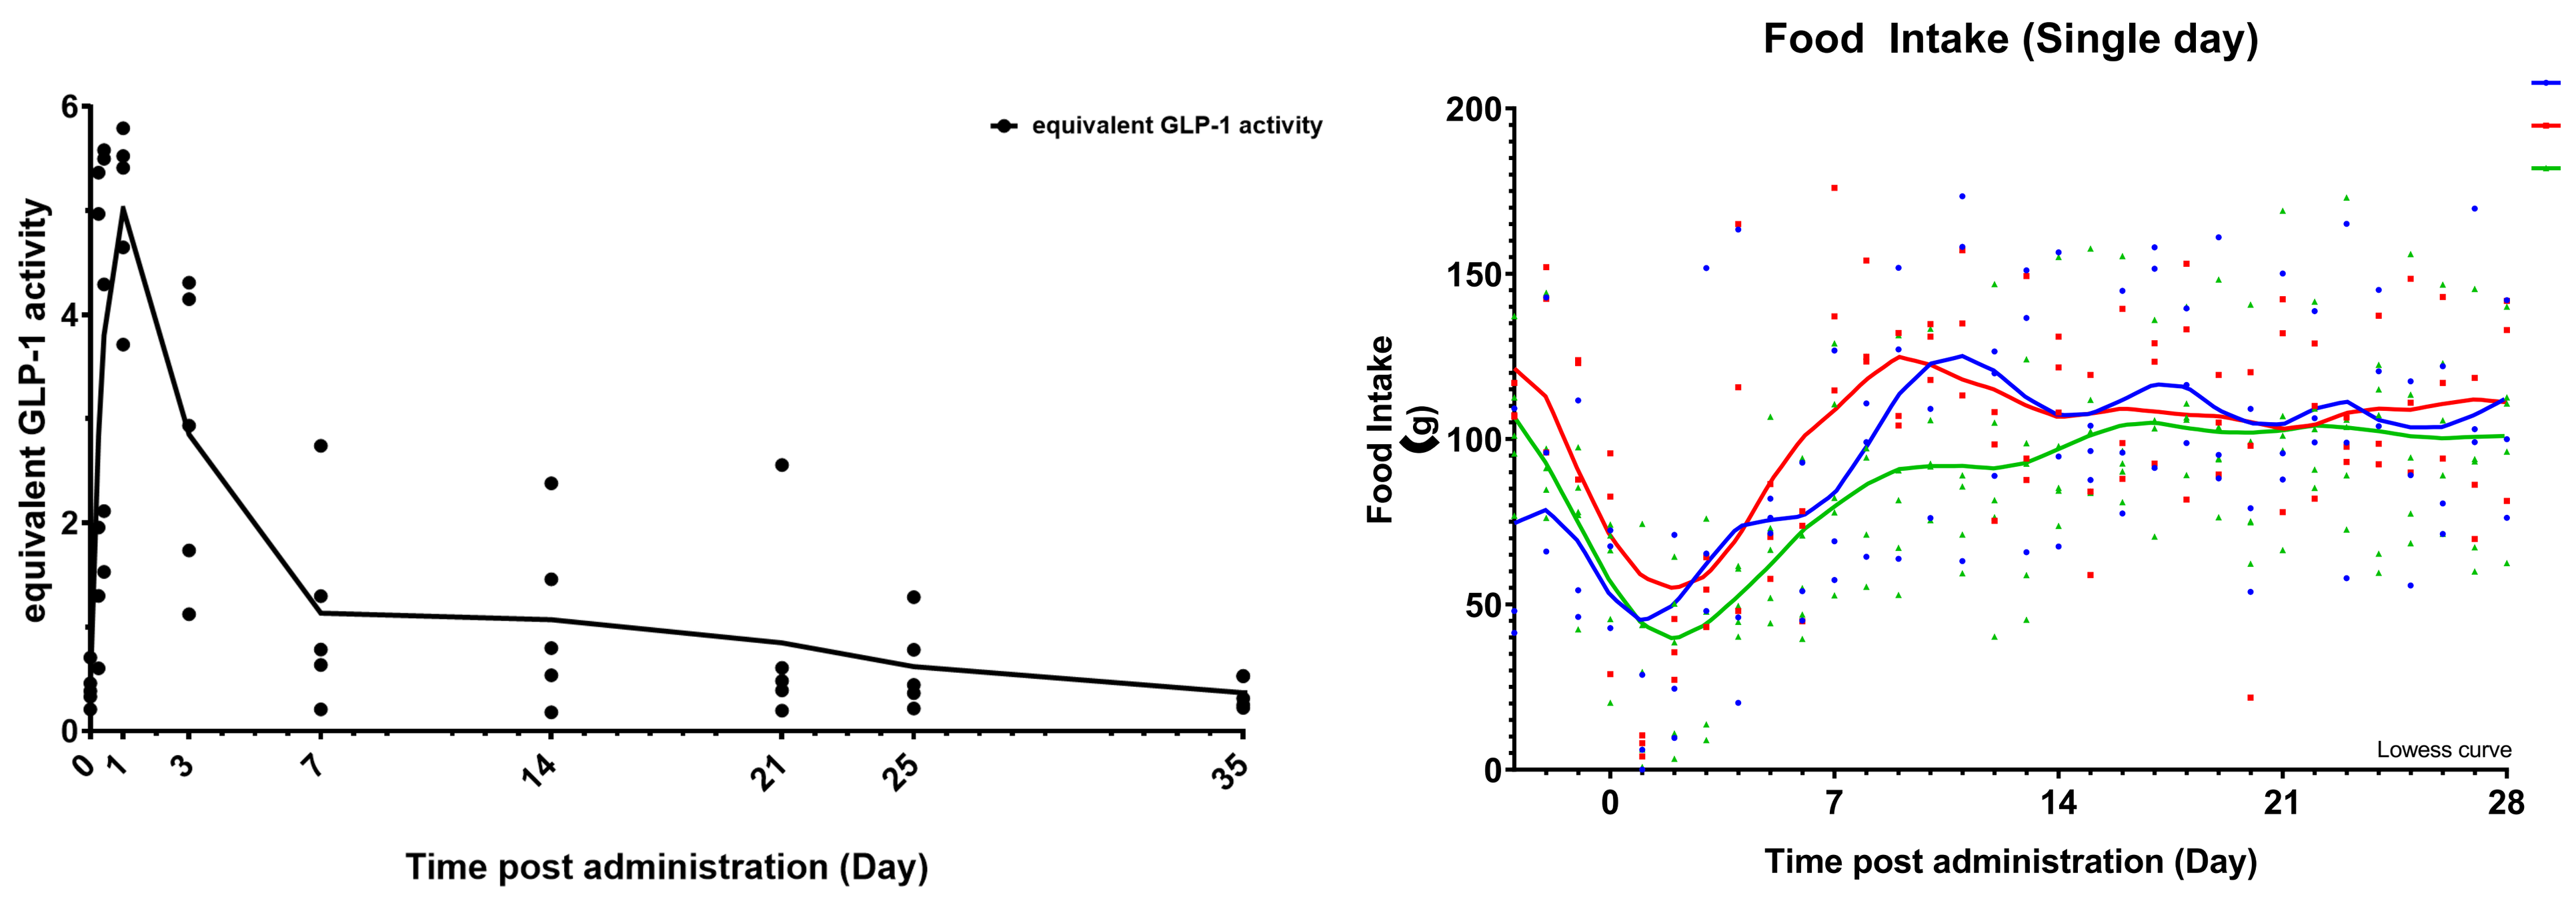
equivalent GLP-1 activity
Time post administration (Day)
Food Intake (Single day)
1st Assay
2nd Assay
3rd Assay
Food Intake (g)
Lowess curve
Time post administration (Day)

Figure 5. Blood samples were collected at various time points and assayed in a GLP-1R/CRE Luciferase Reporter Cell system for GLP-1 equivalent activities (left panel). Daily food consumption values were recorded (right panel).

Pharmacodynamics after multiple HFG1 injections

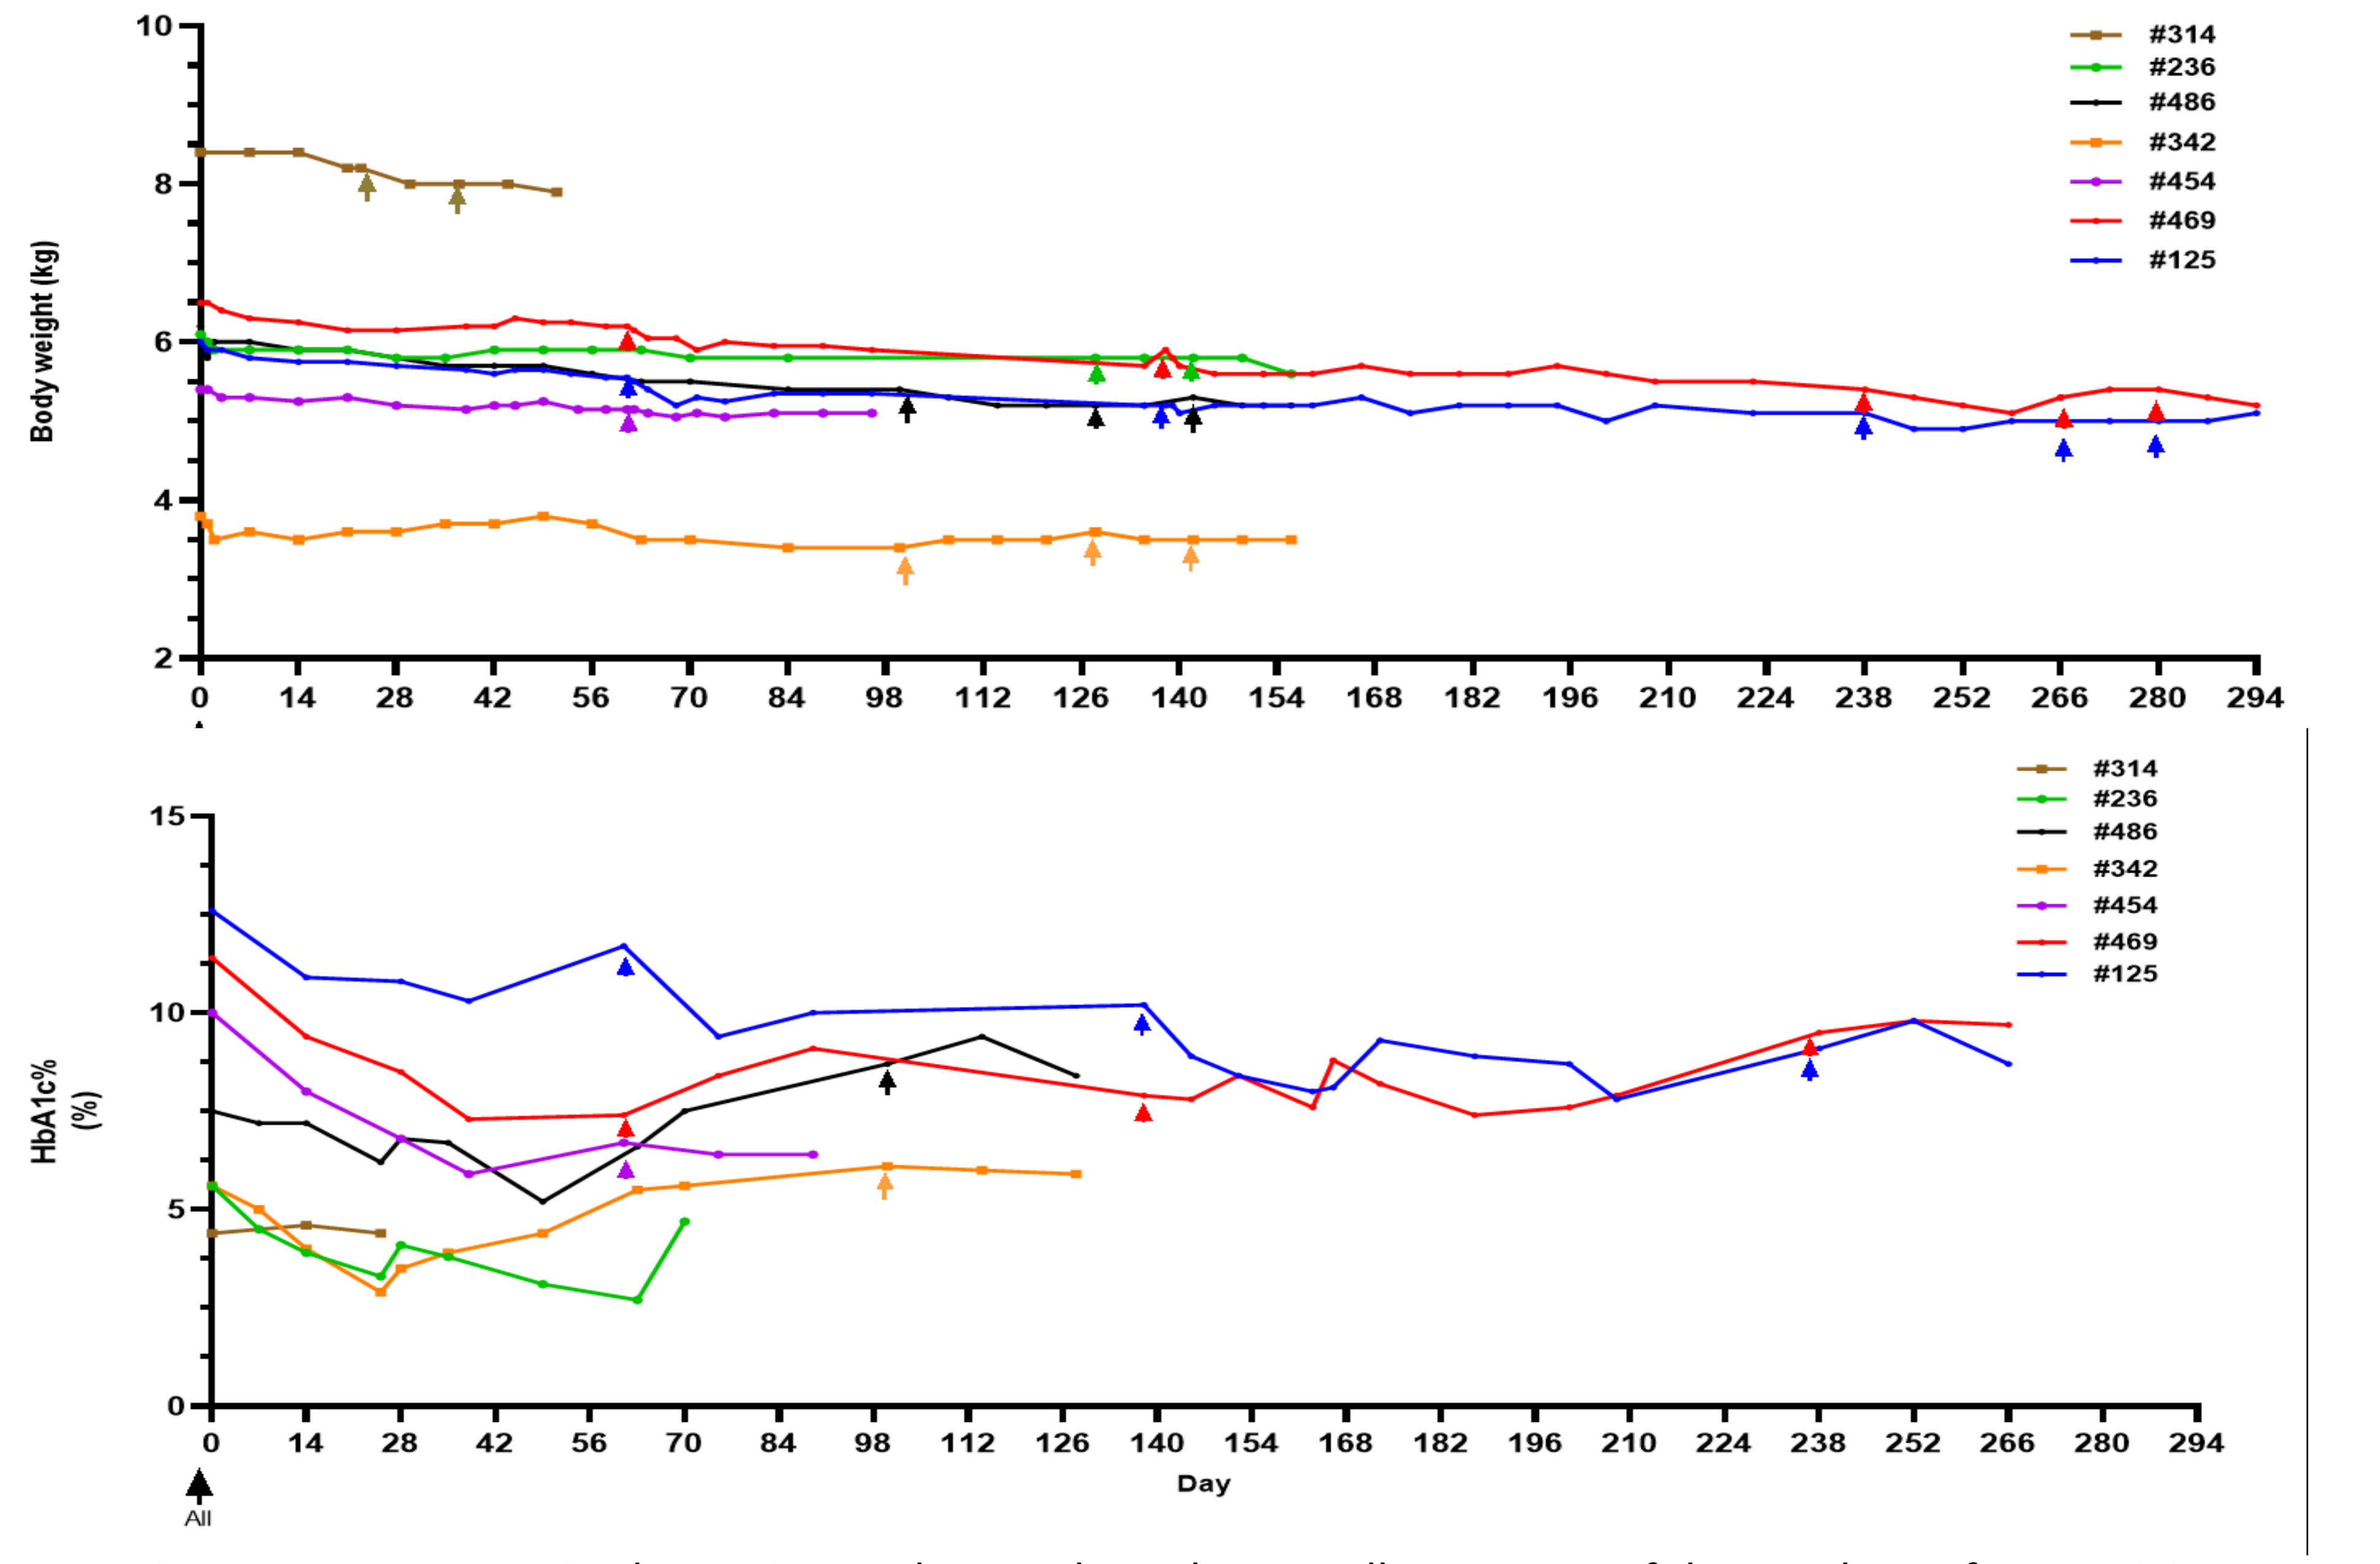
Body weight (kg)
#314
#236
#486
#342
#454
#469
#125
HbA1c% (%)
Day
All

Figure 6. We summarized experiment data to show the overall responses of the monkeys after receiving multiple HFG1 injections. The different animals were color coded and the arrows indicated the time when injections were given.

DISCLOSURES

Authors are employees of Hangzhou Highfield Biopharmaceuticals. For contact: yhxu@hf-biopharm.com